DN101-004: A Multicenter, Open Label, Dose Ranging Study of DN-101 and Docetaxel in Patients with Stage IIIB or IV Non-Small Cell Lung Cancer (NSCLC) after Platinum-Based Chemotherapy

H. West[1], S. Lee[2], J. G. Curd[2], L. Fehrenbacher[3], A. L. Cohn[4], J. O. Hopkins[5], D. H. Irwin[6], D. A. Smith[7], T. E. Boyd[8], M. R. Olsen[9], DN101-004 Study Group;

[1]Swedish Cancer Inst, Seattle, WA; [2]Novacea, Inc., South San Francisco, CA; [3]Kaiser Permanente Med Ctr, Vallejo, CA; [4]Rocky Mountain Cancer Ctr, Denver, CO; [5]Piedmont Hematology Oncology Assoc, Winston-Salem, NC; [6]Alta Bates Comprehensive Cancer Ctr, Berkeley, CA; [7]Northwest Cancer Specialists, P.C., Vancouver, WA; [8]Yakima Valley Memorial Hosp/North Star Lodge, Yakima, WA; [9]Cancer Care Assoc, Tulsa, OK

Objectives

The objective of the study was to determine the maximum tolerated dose (MTD), overall response rate (ORR), progression-free survival (PFS), and overall survival (OS) of DN-101 in combination with docetaxel in advanced NSCLC patients.

Introduction

DN-101

- Dose-intense oral formulation of calcitriol, designed to achieve supra-physiological blood levels of calcitriol without clinically significant hypercalcemia
- Potent vitamin D receptor (VDR) agonist; calcitriol has various anti-neoplastic effects on malignant cells and has synergistic activity when combined with cytotoxic agents, including taxanes
- DN-101 in combination with weekly docetaxel was associated with improved survival in a Phase 2 study in androgen-independent prostate cancer (AIPC); a Phase 3 confirmatory study is underway

We sought to determine whether similar enhancement of the antitumor activity of docetaxel by DN-101 observed in preclinical studies and the Phase 2 study in AIPC would be obtained when combining DN-101 with docetaxel in the treatment of NSCLC.

Patient Population

Key Eligibility Criteria

- Histologically or cytologically proven NSCLC, Stage IIIB or IV, that had progressed on or after platinum-based chemotherapy
- ECOG performance status ≤ 1
- Adequate hematological, liver and renal function
- No more than 1 (DN-101 q21d group) or 2 (DN-101 q7d group) prior chemotherapy regimens for NSCLC
- No prior docetaxel therapy

Methods

Study Design and Treatment

Multi-center, open-label, dose ranging study of docetaxel in combination with DN-101

Treatment Groups:

- DN-101 q21d – Sequential escalation of DN-101 administered on day 1 at doses at 45 (n=5), 75 (n=4), 135 (n=3) or 180 µg (n=53)
- DN-101 q7d – Sequential escalation of DN-101 administered on day 1 (180 µg) followed by 45 (n=5), 90 (n=4) or 180 µg (n=12) on days 8 and 15

Docetaxel (75 mg/m^2) was given q21d on day 2 for all patients

Methods *(continued)*

A minimum of 3 patients were required to complete cycle 1 without dose-limiting toxicity (DLT) prior to treating patients in the next higher DN-101 dose level. Enrollment in the highest tolerated DN-101 q21d dose group was to be expanded to up to 50 patients. Treatment cycles were repeated at the same dose level until disease progression or unacceptable toxicity.

Assessments

Response and progression were evaluated using Response Evaluation Criteria in Solid Tumors (RECIST) every 6 weeks.

Data Analysis

Baseline characteristics and results were combined for individual dose cohorts within each treatment group.

Analysis Populations

- Efficacy Evaluable Population — received at least one dose of study treatment (DN-101 or docetaxel) and had at least one valid post-baseline tumor assessment
- Safety Population — received at least one dose of study treatment (DN-101 or docetaxel)

Results

Table 1. Baseline Characteristics

	DN-101 q21d (n=65)	DN-101 q7d (n=21)	Overall (n=86)
Age, median, y (range)	62.8 (34 – 86)	65.4 (53 – 80)	63.2 (34 – 86)
Gender (%)			
Male	30 (46.2)	13 (61.9)	43 (50.0)
Female	35 (53.8)	8 (38.1)	43 (50.0)
ECOG (%)			
0	23 (35.4)	7 (33.3)	30 (34.9)
1	40 (61.5)	13 (61.9)	53 (61.6)
2	1 (1.5)	1 (4.8)	2 (2.3)
Lung Cancer Histology (%)			
Adenocarcinoma	41 (63.1)	13 (61.9)	54 (63.8)
Squamous cell carcinoma	9 (13.9)	1 (4.8)	10 (11.6)
Other	15 (23.1)	7 (33.3)	22 (25.6)
Lung Cancer Stage (%)			
IIIA	1 (1.5)	0	1 (1.2)
IIIB	5 (7.7)	1 (4.8)	6 (7.0)
IV	59 (90.8)	20 (95.2)	79 (91.9)
Prior Lung Cancer Therapy (%)*			
Cisplatin + paclitaxel	2 (3.1)	0	2 (2.3)
Cisplatin + gemcitabine	2 (3.1)	2 (9.5)	4 (4.7)
Carboplatin + paclitaxel	52 (80.0)	15 (71.4)	67 (77.9)
Other	20 (30.8)	10 (47.6)	30 (34.9)

*Includes first-line or second-line therapy

Results *(continued)*

Table 2. Exposure

	DN-101 q21d (n=65)	DN-101 q7d (n=21)
Number of cycles		
Median (range)	4 (1 – 22)	2 (1 – 10)
Cumulative DN-101 dose (µg)		
Median (range)	540 (90 – 3375)	1080 (180 – 5580)
Cumulative docetaxel dose (mg/m^2)		
Median (range)	228.6 (72 – 1468)	146.8 (74 – 671)

Table 3. Efficacy

	DN-101 q21d (n=61)	DN-101 q7d (n=17)
Tumor Response Rate (PR & CR)	6.6%	5.9%
Disease Stabilization Rate (PR, CR & SD)	60.7%	58.8%
Best Overall Response (%)		
Confirmed PR	4 (6.6)	1 (5.9)
Unconfirmed PR	2 (3.3)	1 (5.9)
SD	31 (50.8)	8 (47.1)
PD	24 (39.3)	7 (41.2)
Median Progression-Free Survival (weeks) (95% CI)	11.6 (8.4, 17.4)	14.1 (6.0, 20.0)
12-month Survival Rate (95% CI)	30.5 (18.9, 42.2)	40.0 (15.2, 64.8)
Median Survival (months) (95% CI)	6.9 (5.5, 9.7)	8.8 (7.1, NA)

Kaplan-Meier Curve for Overall Survival by DN-101 Regimen

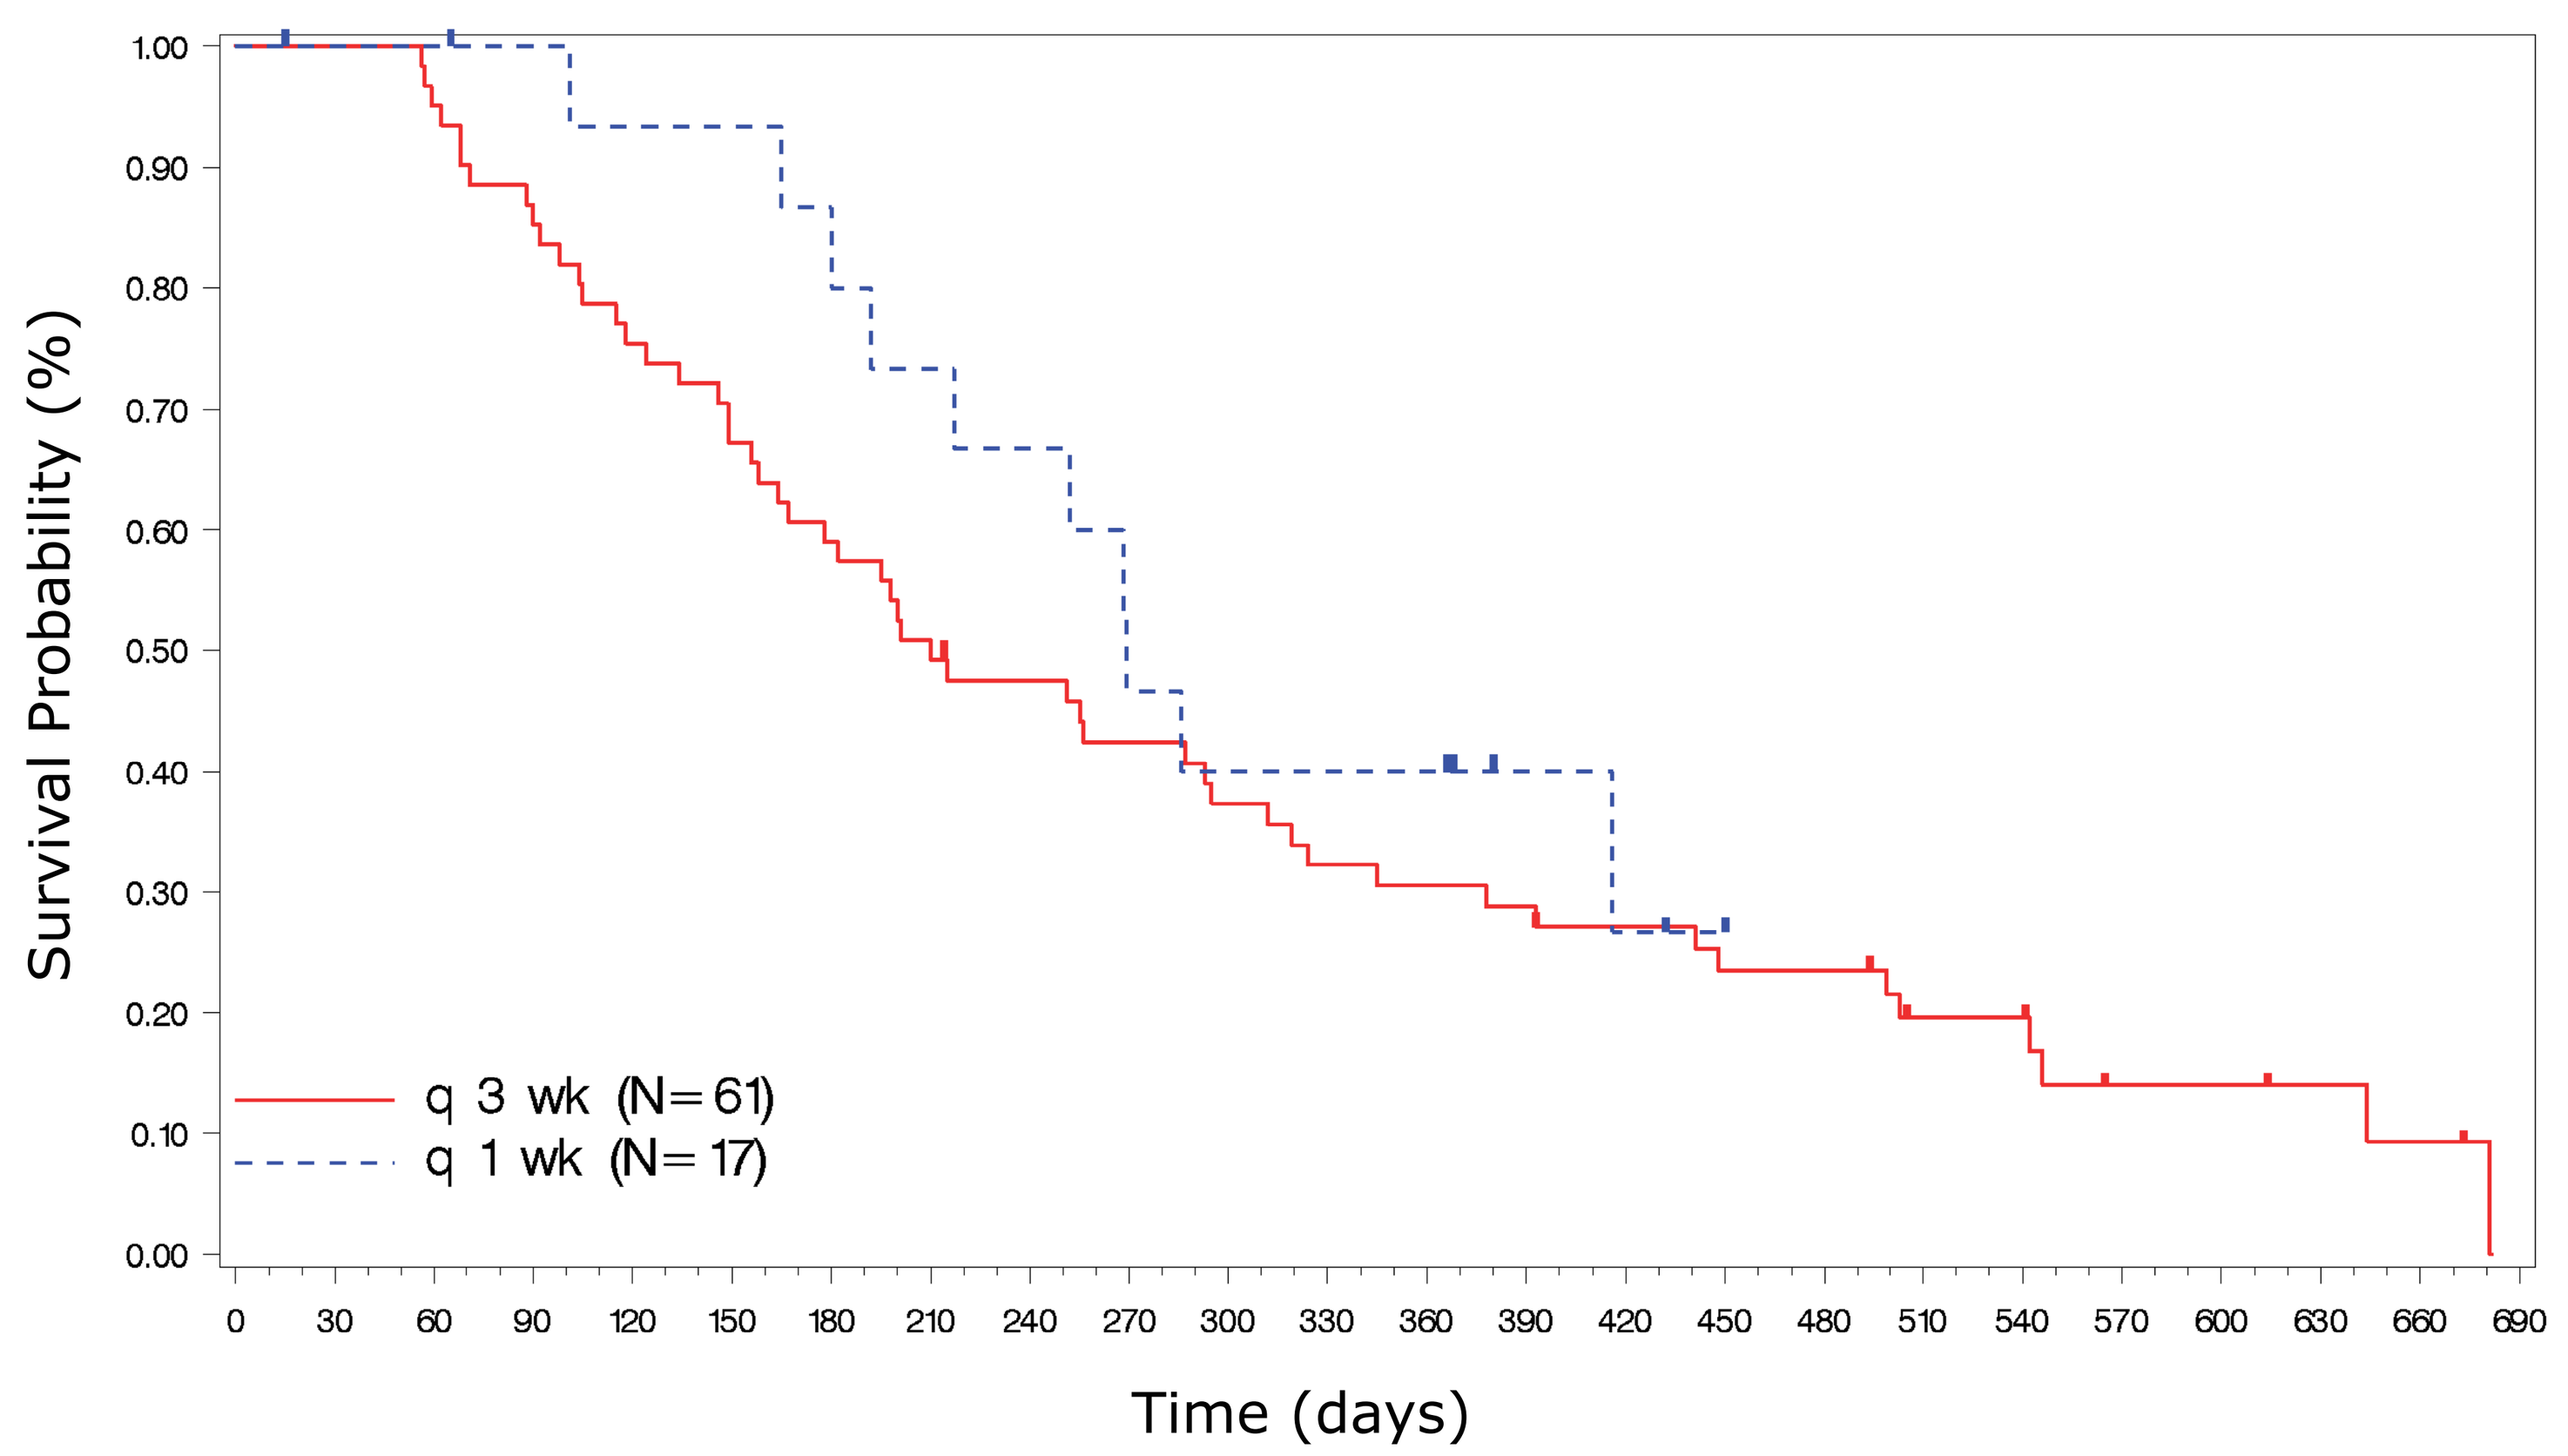
Survival Probability (%)
1.00
0.90
0.80
0.70
0.60
0.50
0.40
0.30
0.20
0.10
0.00
q 3 wk (N=61)
q 1 wk (N=17)
0 30 60 90 120 150 180 210 240 270 300 330 360 390 420 450 480 510 540 570 600 630 660 690
Time (days)

Results *(continued)*

Table 4. Adverse Events

	DN-101 q21d n=65 (%)	DN-101 q7d n=21 (%)	Overall n=86 (%)
Neutropenia			
Any	41 (63.1)	11 (52.4)	52 (60.5)
Grade 3/4	36 (55.4)	11 (52.4)	47 (54.7)
Leukopenia			
Any	17 (26.2)	2 (9.5)	19 (22.1)
Grade 3/4	12 (18.5)	1 (4.8)	13 (15.1)
Febrile neutropenia	4 (6.2)	1 (4.8)	5 (5.8)
Asthenia			
Any	8 (12.3)	1 (4.8)	9 (10.5)
Grade 3/4	4 (6.2)	1 (4.8)	5 (5.8)
Fatigue			
Any	39 (60.0)	7 (33.3)	46 (53.5)
Grade 3/4	5 (7.7)	0	5 (5.8)
Stomatitis			
Any	6 (9.2)	0	6 (7.0)
Grade 3/4	0	0	0
Hypercalcemia			
Any	6 (9.2)	6 (28.6)	12 (14.0)
Grade 3/4	0	2 (9.5)	2 (2.3)

Three fatal AEs that were considered unrelated to DN-101
- 2 on DN-101 q21d (lung infiltration, pneumonitis)
- 1 on DN-101 q7d (intestinal perforation)

Conclusion

- **MTD of DN-101 was not reached; DN-101 q21d and q7d up to 180 µg in combination with docetaxel q21d was well-tolerated**
- **No unexpected toxicities were reported; grade 3/4 toxicities and fatal AEs were consistent with those reported for docetaxel alone**
- **Stomatitis (9%, q21d; 0%, q7d) and grade 3/4 asthenia (6%, q21d; 5%, q7d) were less frequently reported for DN-101 combined with docetaxel as compared to published reports of docetaxel alone (26% and 18%, respectively)[1]**
- **Efficacy of DN-101 plus docetaxel appeared comparable to (q21d) or better than (q7d) that reported for docetaxel alone[2,3]**
- **Overall results, including possible improvement in toxicity, support further investigation of DN-101 in NSCLC**

References

1 sanofi-aventis U.S. Taxotere® (docetaxel) Injection Concentrate. Prescribing Information. October 16, 2006.

2 Shepherd FA et al. Prospective randomized trial of docetaxel versus best supportive care in patients with non-small-cell lung cancer previously treated with platinum-based chemotherapy. J Clin Oncol 2000; 18:2095-2103.

3 Fossella FV et al. Randomized Phase III trial of docetaxel versus vinorelbine or ifosfamide in patients with advanced non-small-cell lung cancer previously treated with platinum-containing chemotherapy regimens. J Clin Oncol 2000; 18:2354-62.

Acknowledgement & Disclosures

Supported by Novacea, Inc. John G. Curd is an employee of Novacea, Inc. S. Lee was an employee of Novacea, Inc.

2007 ASCO Annual Meeting, June 1-4, 2007, Chicago, IL